Phase I Trials: Model-based Assessment to Identify a Clinical Relevant Change in Heart Rate

Martin Fink[1], Olivier Luttringer[1], Peter Gergely[2], Erik Wallstroem[2], Michael Looby[1] and Goonaseelan Pillai[1]

[1]Modeling & Simulation, Novartis Pharma AG, Basel, Switzerland and [2]Translational Medicine, Novartis Pharma AG, Basel, Switzerland

Objectives

- The primary objective of this analysis was to ascertain, through an integrated PK/PD model-based approach, what measure of change in heart rate (HR) should be considered as clinically relevant in phase I trials. Interindividual and between-study variability of the circadian variations of HR demand a more sophisticated approach than simple baseline-correction, when attempting to distinguish drug effects from usual changes in HR.

Methods

- Placebo and predose hourly average HR data from 24-h holter monitoring from seven phase I clinical studies were pooled (n=405, >700 full days of recordings). The basic mathematical model1 consisted of a sum of five cosine functions to replicate the circadian variations in (with periods p_k of 24, 12, 8, 6, 4.8 hours, respectively).

$$H = H_{MES} + \sum_{k=1}^{5} a_k \cos\left(\frac{2\pi}{p_k}(t-\tau_k)\right),$$

 where H denotes HR, H_{MES} the mesor or mean HR, a_k the amplitude, p_k the period and τ_k the phase shift of the kth cosine function.
- Interindividual variability (IIV) was assessed for H_{MES} and a_k using a log-normal distribution. An example is given for the notional structural model parameter P,

$$P_i = TVP\exp(\eta_{P,i}),$$

 whereas IIV for the phase shift τ_k was found to be normally distributed

$$\tau_{k,i} = TV\tau_k + \eta_{\tau_k,i}.$$

- Residual variability was tested as additive or proportional or both.
- Study, sex, and weight were tested during the covariate building of a non-linear mixed effects model as well as the placebo effect compared to predose. Covariates were modeled similarly as the IIV with $\exp(\beta_P)$ for H_{MES} and a_k and additive $+\beta_P$ for τ_k.
- Covariates were first investigated for only the predose data and later for changes between predose and placebo during the treatment period.
- Statistical shrinkage in empirical Bayes estimates (EBEs) of model parameters used for diagnostic purposes was evaluated as

$$sh_P = 1 - \frac{SD(\eta_{EBE,P})}{\varpi_P},$$

 where sh_P is shrinkage in model parameter P, $SD(\eta_{EBE,P})$ is the standard deviation of the individual EBEs of IIV in parameter P, and ω_P is the model estimate of the standard deviation in the IIV associated with parameter P.

Results

- Due to the high number of possible covariates (7 studies + 2 genders + weight)*(11 parameters) we applied several independent methods to find the optimal covariate model for the predose HR data. Monolix3.1[2] was able to run a full covariate model and did not find a significant effect of weight (results see **Table 1**). Xpose4[3] was used to fit a generalized additive model (GAM), which suggested very similar covariates as found with the Monolix full covariate model. Nonmem VI[4] showed large difficulties fitting the large model such that a stepwise covariate search was not possible with the full data set (analysis of a subset of data using studies 1, 2, and 3 led to equivalent covariates as presented in **Table 1**).

Table 1. *Covariates with full covariate model in Monolix and GAM using Xpose with ST1 as the reference*

	Sex	ST2	ST3	ST4	ST5	ST6	ST7
H_{MES}	p*	c	c*	c	c*	c	c
a1	p*	c*		p*		p*	p*
τ1		p*	p*	p*	p*	p*	
a2				p*	p*	p*	p*
τ2	p*	p*	c*	p*	c*		p*
a3		*	*	p*	p*		p*
τ3		*	*	*	*	*	
a4	p*	p*	p*	p*		p	*
τ4	p*	p*	p*				p*
a5							p
τ5			p			p	p

*p=statistically significant with p<0.05; c=clinically relevant change (>1 bpm or >0.5 hours), but not significant at p<0.05; *=suggested by GAM (a5, τ5 not tested)*

- The model fit to the HR placebo data using the five cosine functions was very good (see **Figures 1 and 2**). Relative standard errors (r.s.e.) for the fixed effects were below 10% (except 16% for AP5), whereas the r.s.e. for significant covariates was up to 38%. Shrinkage of the individual parameter estimates of H_{MES}, *a1, τ1, a2, τ2, a3, τ3, a4, τ4, a5,* and *τ5* was 2%, 28%, 34%, 50%, 54%, 68%, 100%, 77%, 60%, 100%, and 100%, respectively. The log-likelihood test and Akaike Information Criterion both supported the use of five cosine functions.

Figure 1. *Goodness-of-fit plots for predose fit from Monolix.*

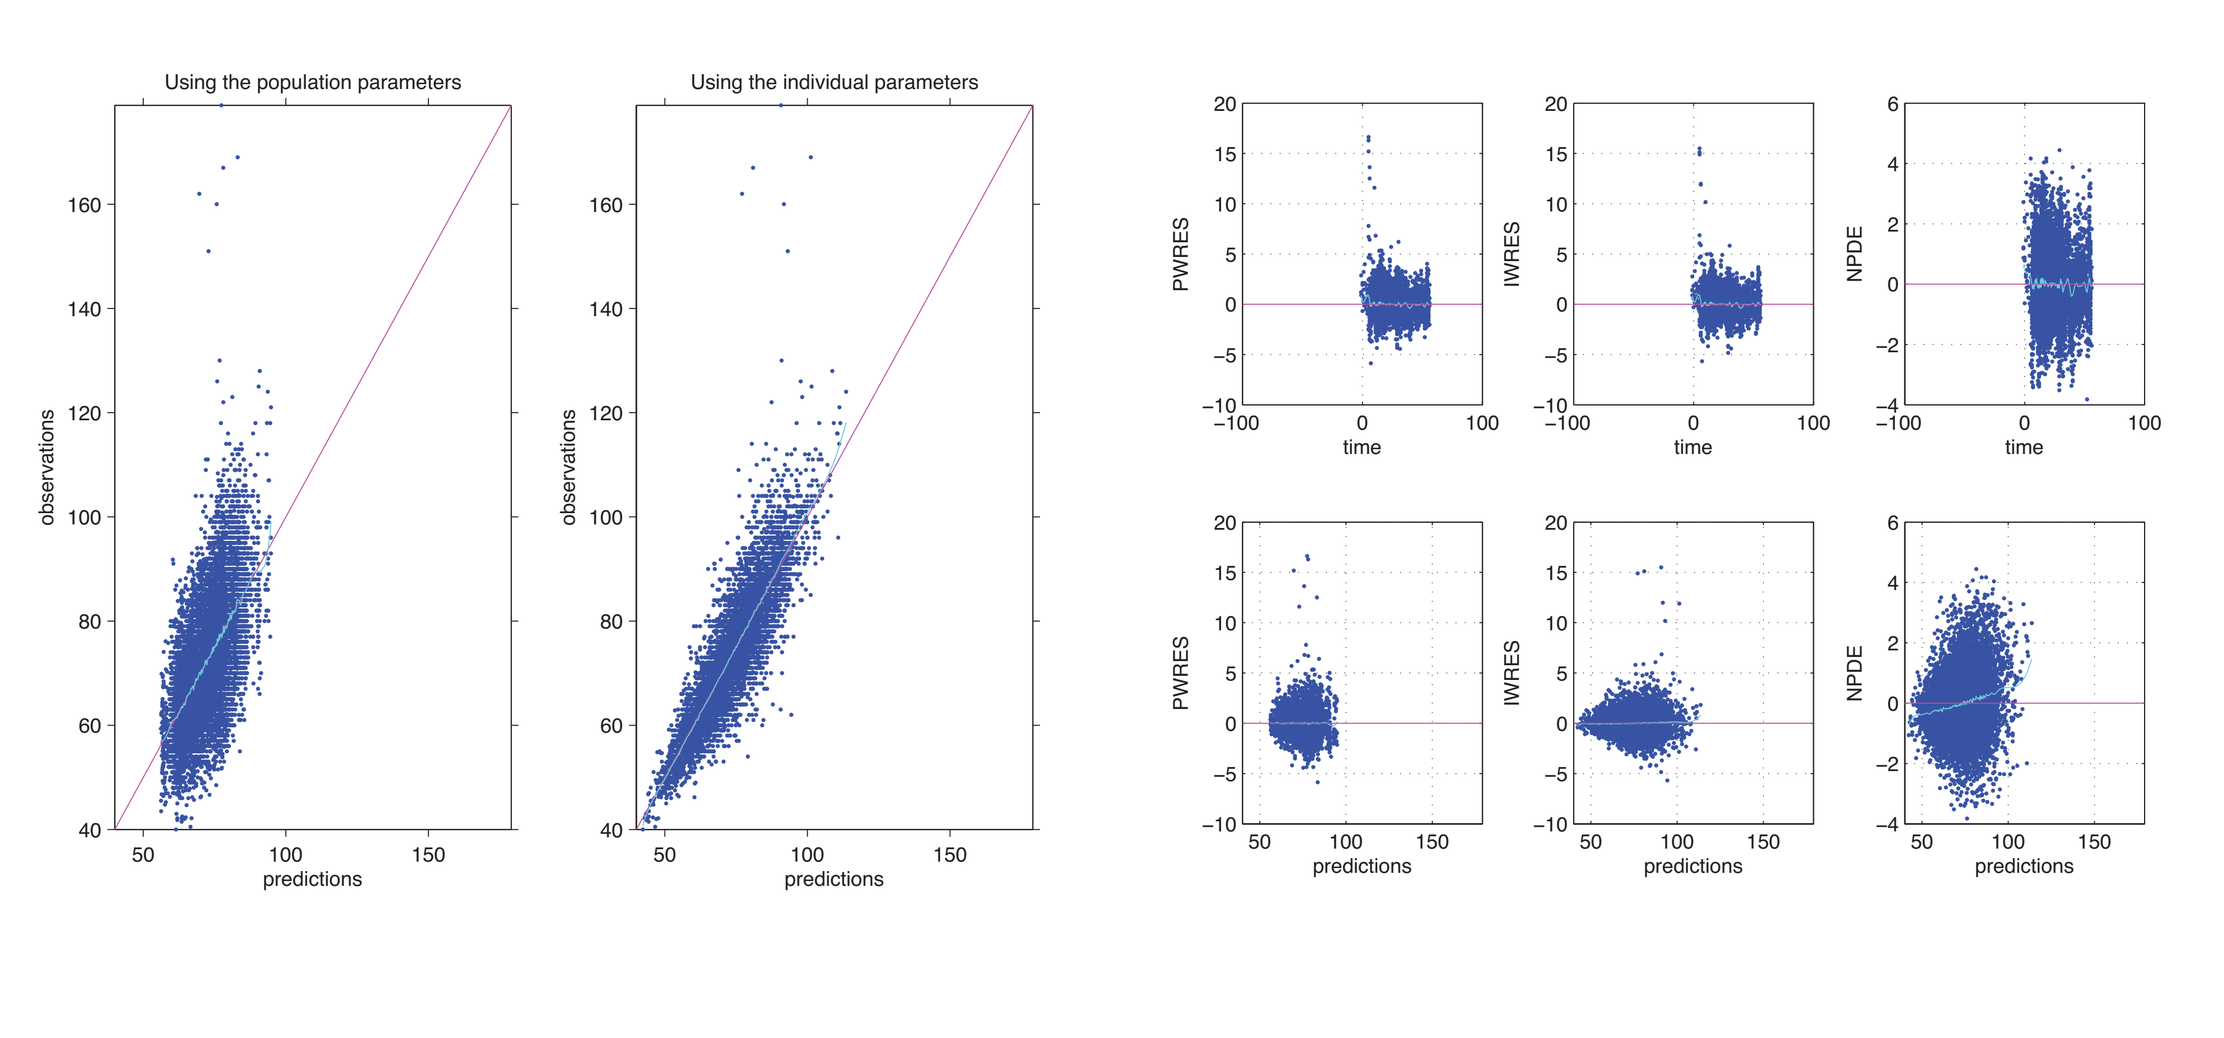

Figure 2. *Randomly selected individual fits of predose HR values*

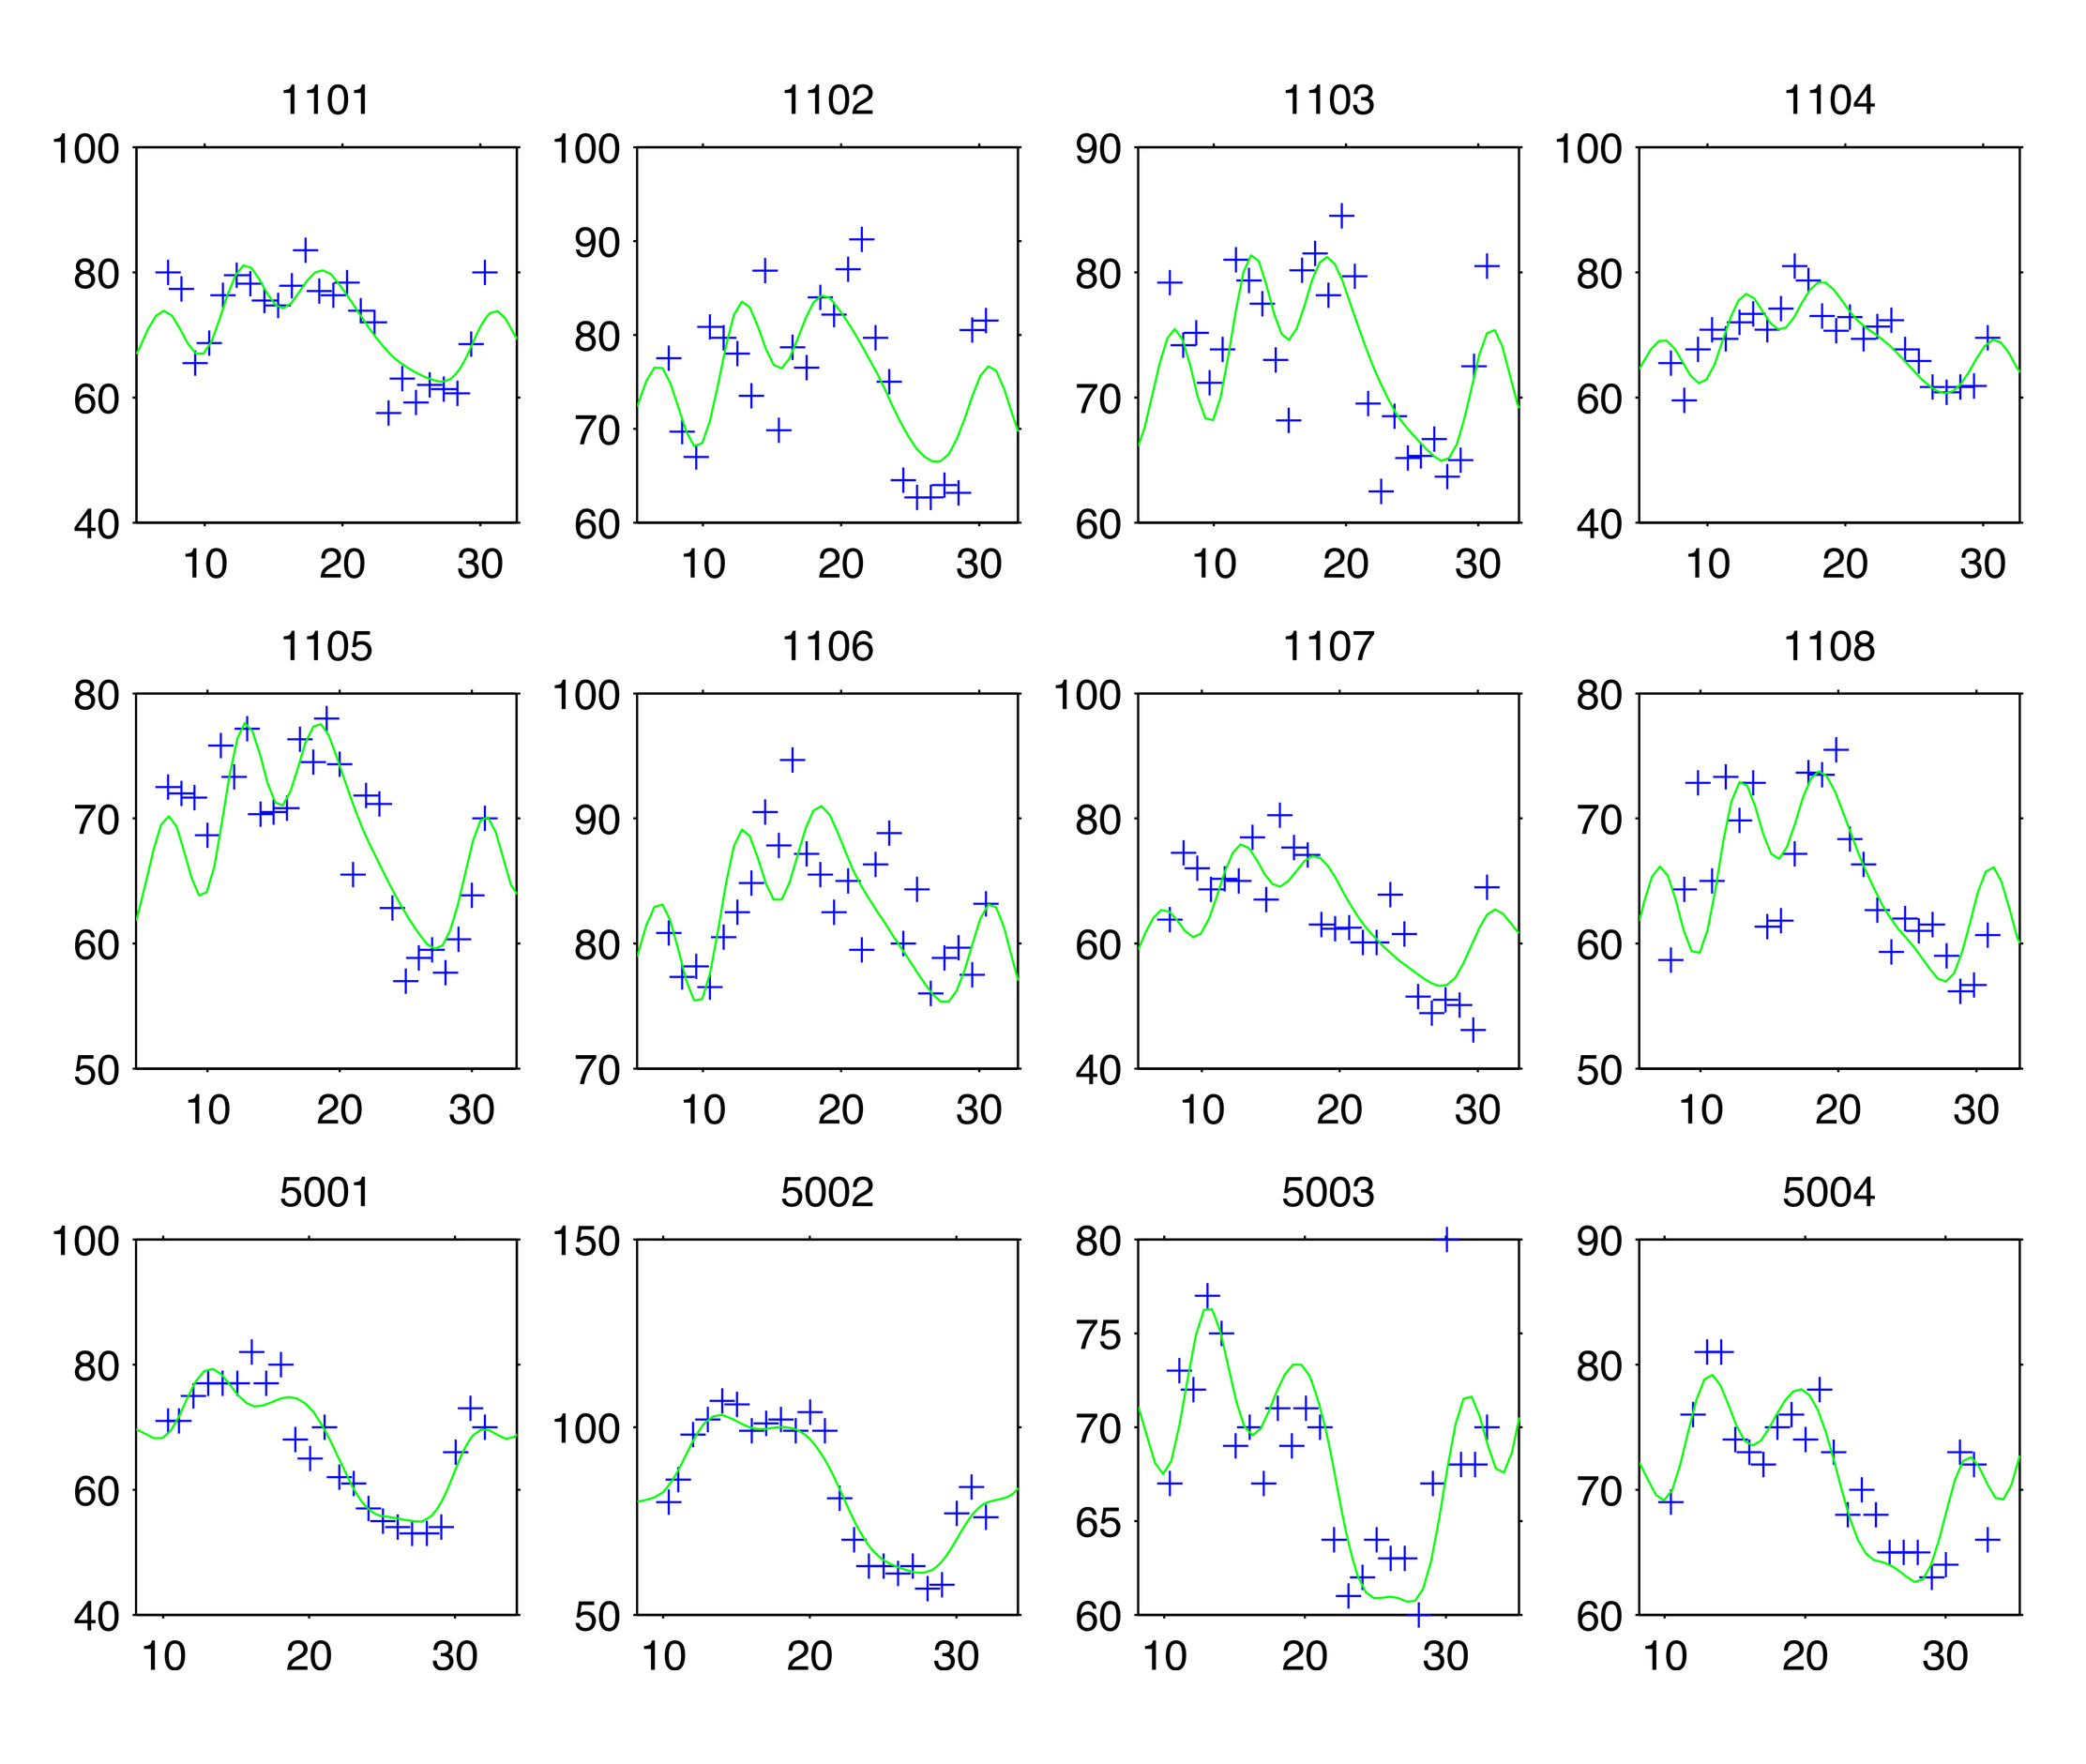

- Typical HR changes over the day were significantly different between studies (see **Figure 3**). HR over the day ranged from approximately 60 to 80 bpm (in a typical male subject). Gender differences could be found for the mesor (~6.6% or ~5 bpm higher for a female subject), but no statistically significant study dependence was noted even though the covariates for the studies implied differences of 1.5–3.2 bpm. The placebo effect on the mesor was always smaller than this gender difference (0.8–6.3% increase).

Figure 3. *Typical circadian HR variation for males in the investigated studies and gender difference represented using study 1*

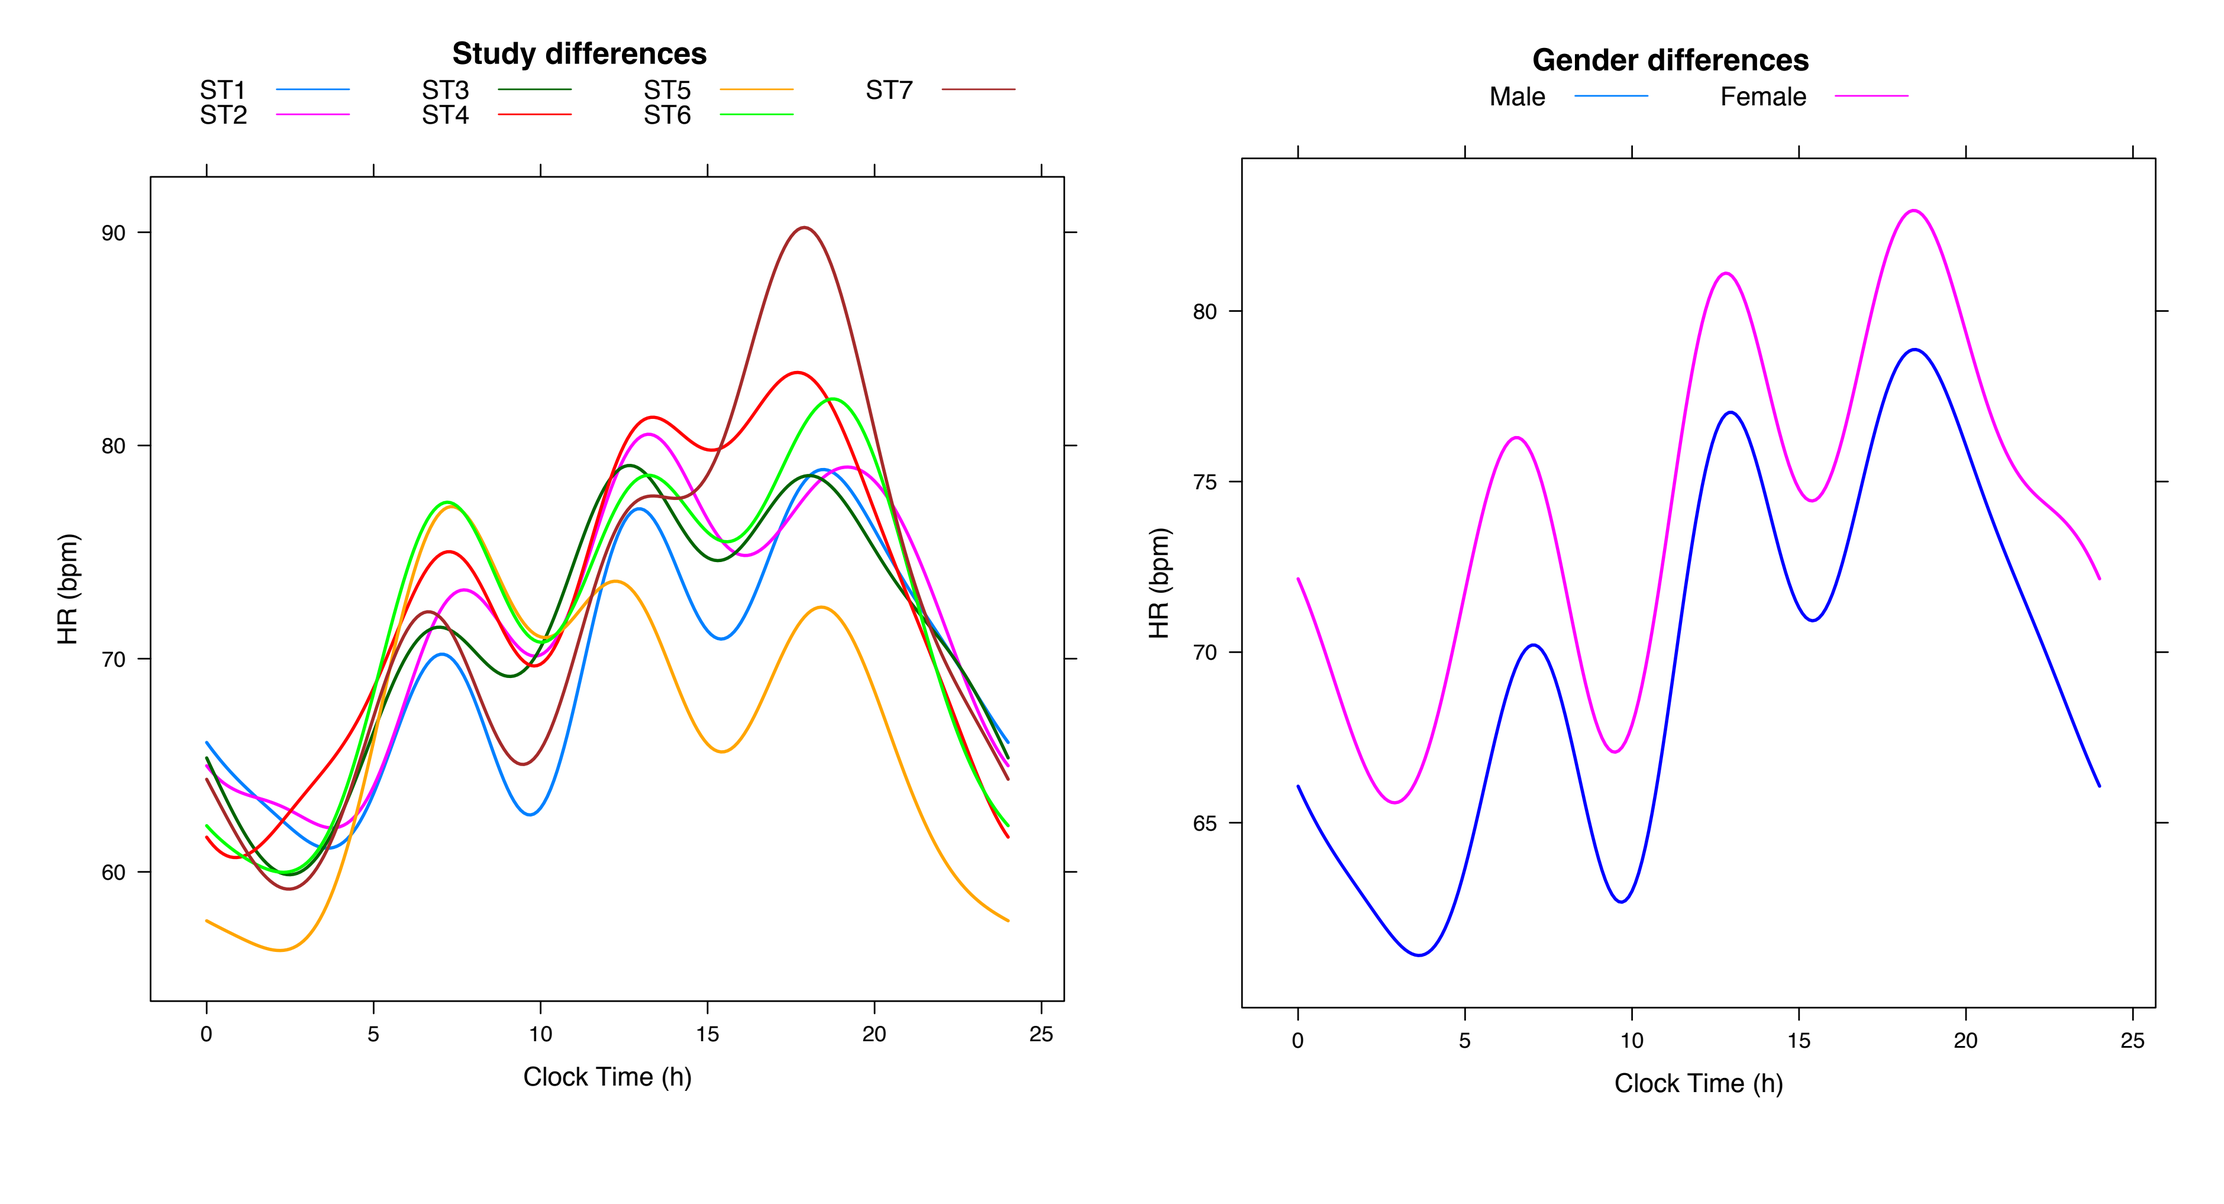
Study differences
ST1
ST2
ST3
ST4
ST5
ST6
ST7
Gender differences
Male
Female
HR (bpm)
Clock Time (h)

- Study dependence was found on the phase shifts and on the amplitudes of the cosine functions, and the latter in general denoted for changes of less than 2 bpm for the typical subjects. Nonetheless, in combination the differences in amplitudes and phase shifts lead to a maximum time-wise difference between studies of up to 18.9 bpm and a maximal difference between genders of more than 11% (or ~8 bpm) in predose data. Note that the largest between-study variability occurs in afternoon and evening.
- The overall variability in the data can be seen in **Figure 4**, showing the predose data values with the typical HR changes in male subjects.

Figure 4. *Variability in predose data compared to typical response*

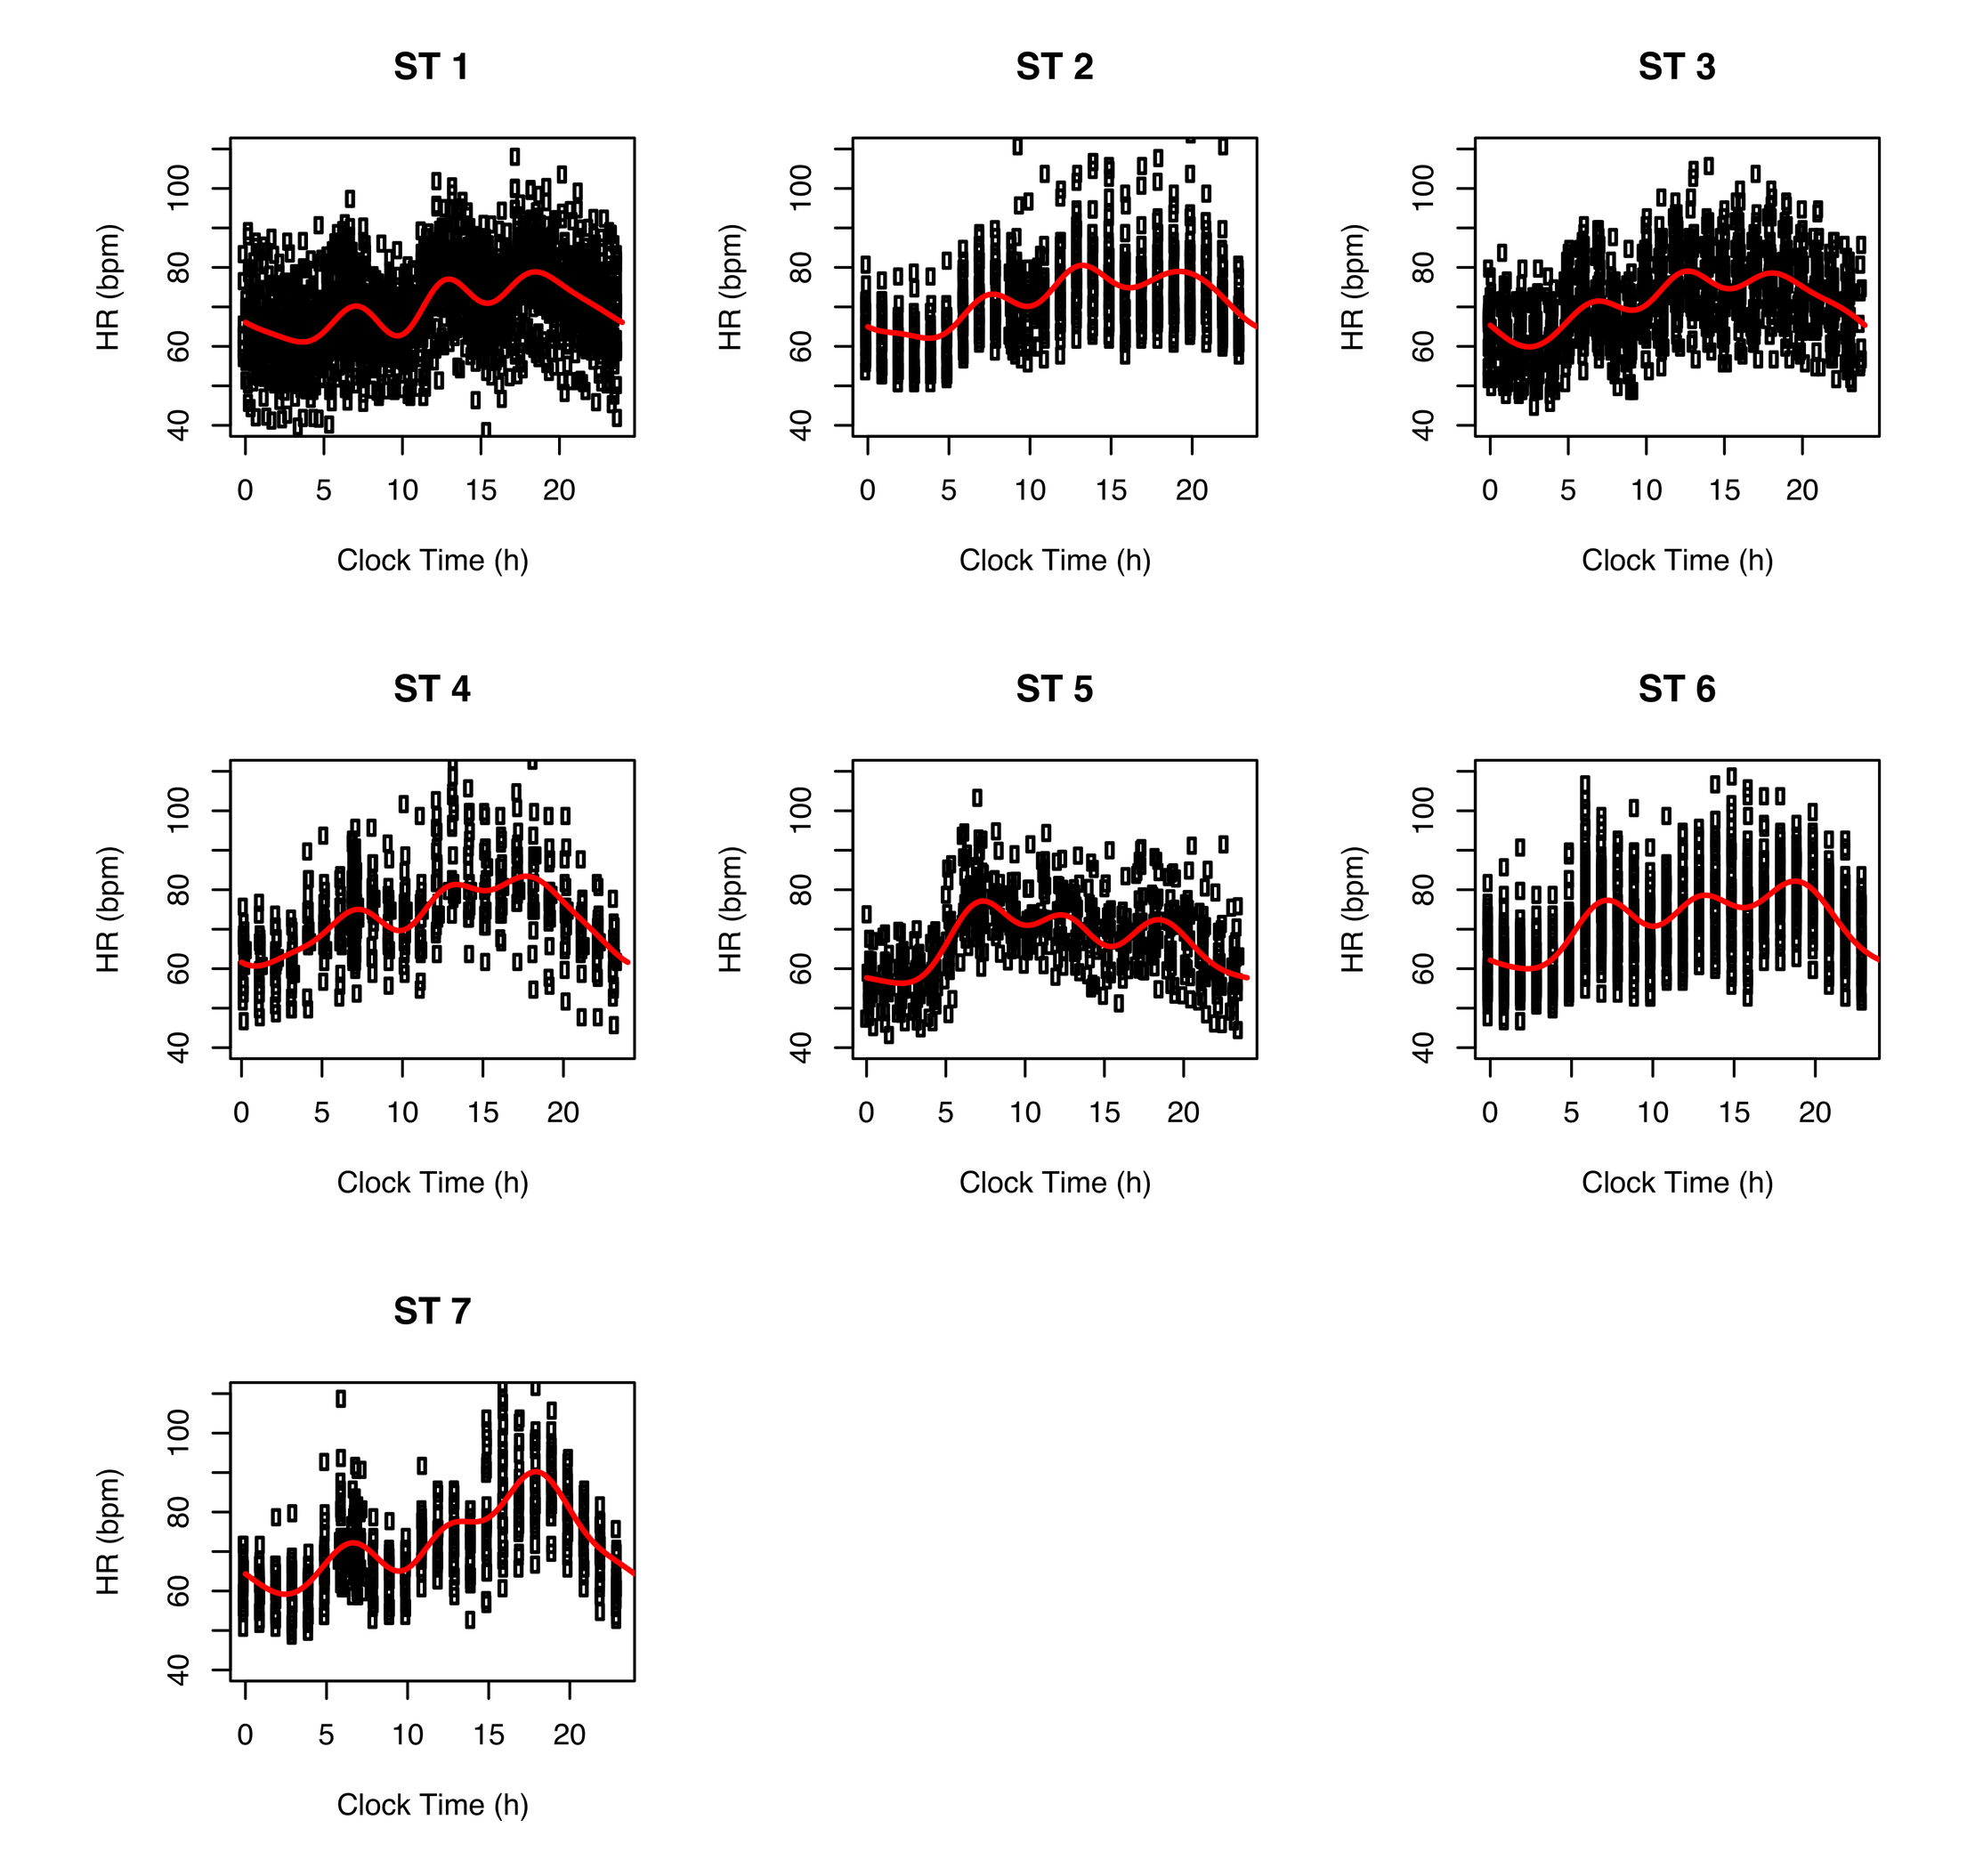
ST 1
ST 2
ST 3
ST 4
ST 5
ST 6
ST 7
HR (bpm)
Clock Time (h)

- The treatment effect in the placebo group was mainly investigated on the mean HR (or mesor). Using inter-occasion-variability for each study day one can see (**Figure 5**) a slight increase in the mesor from days -1 and 0 (predose) to days 1 and later (treatment). The actual increase was occurred either within the first treatment days or more gradually and was very study dependent. Using a stepwise increase between predose and treatment the placebo effect was estimated to be 0.8–6.3% as mentioned before. Estimation of the linearized Fisher information matrix was not possible due to software memory capacities of Monolix3.1, therefore no standard errors or log-likelihood estimates could be derived for the full model.

Figure 5. *Changes in mean HR for different study days per study (using IOV per day)*

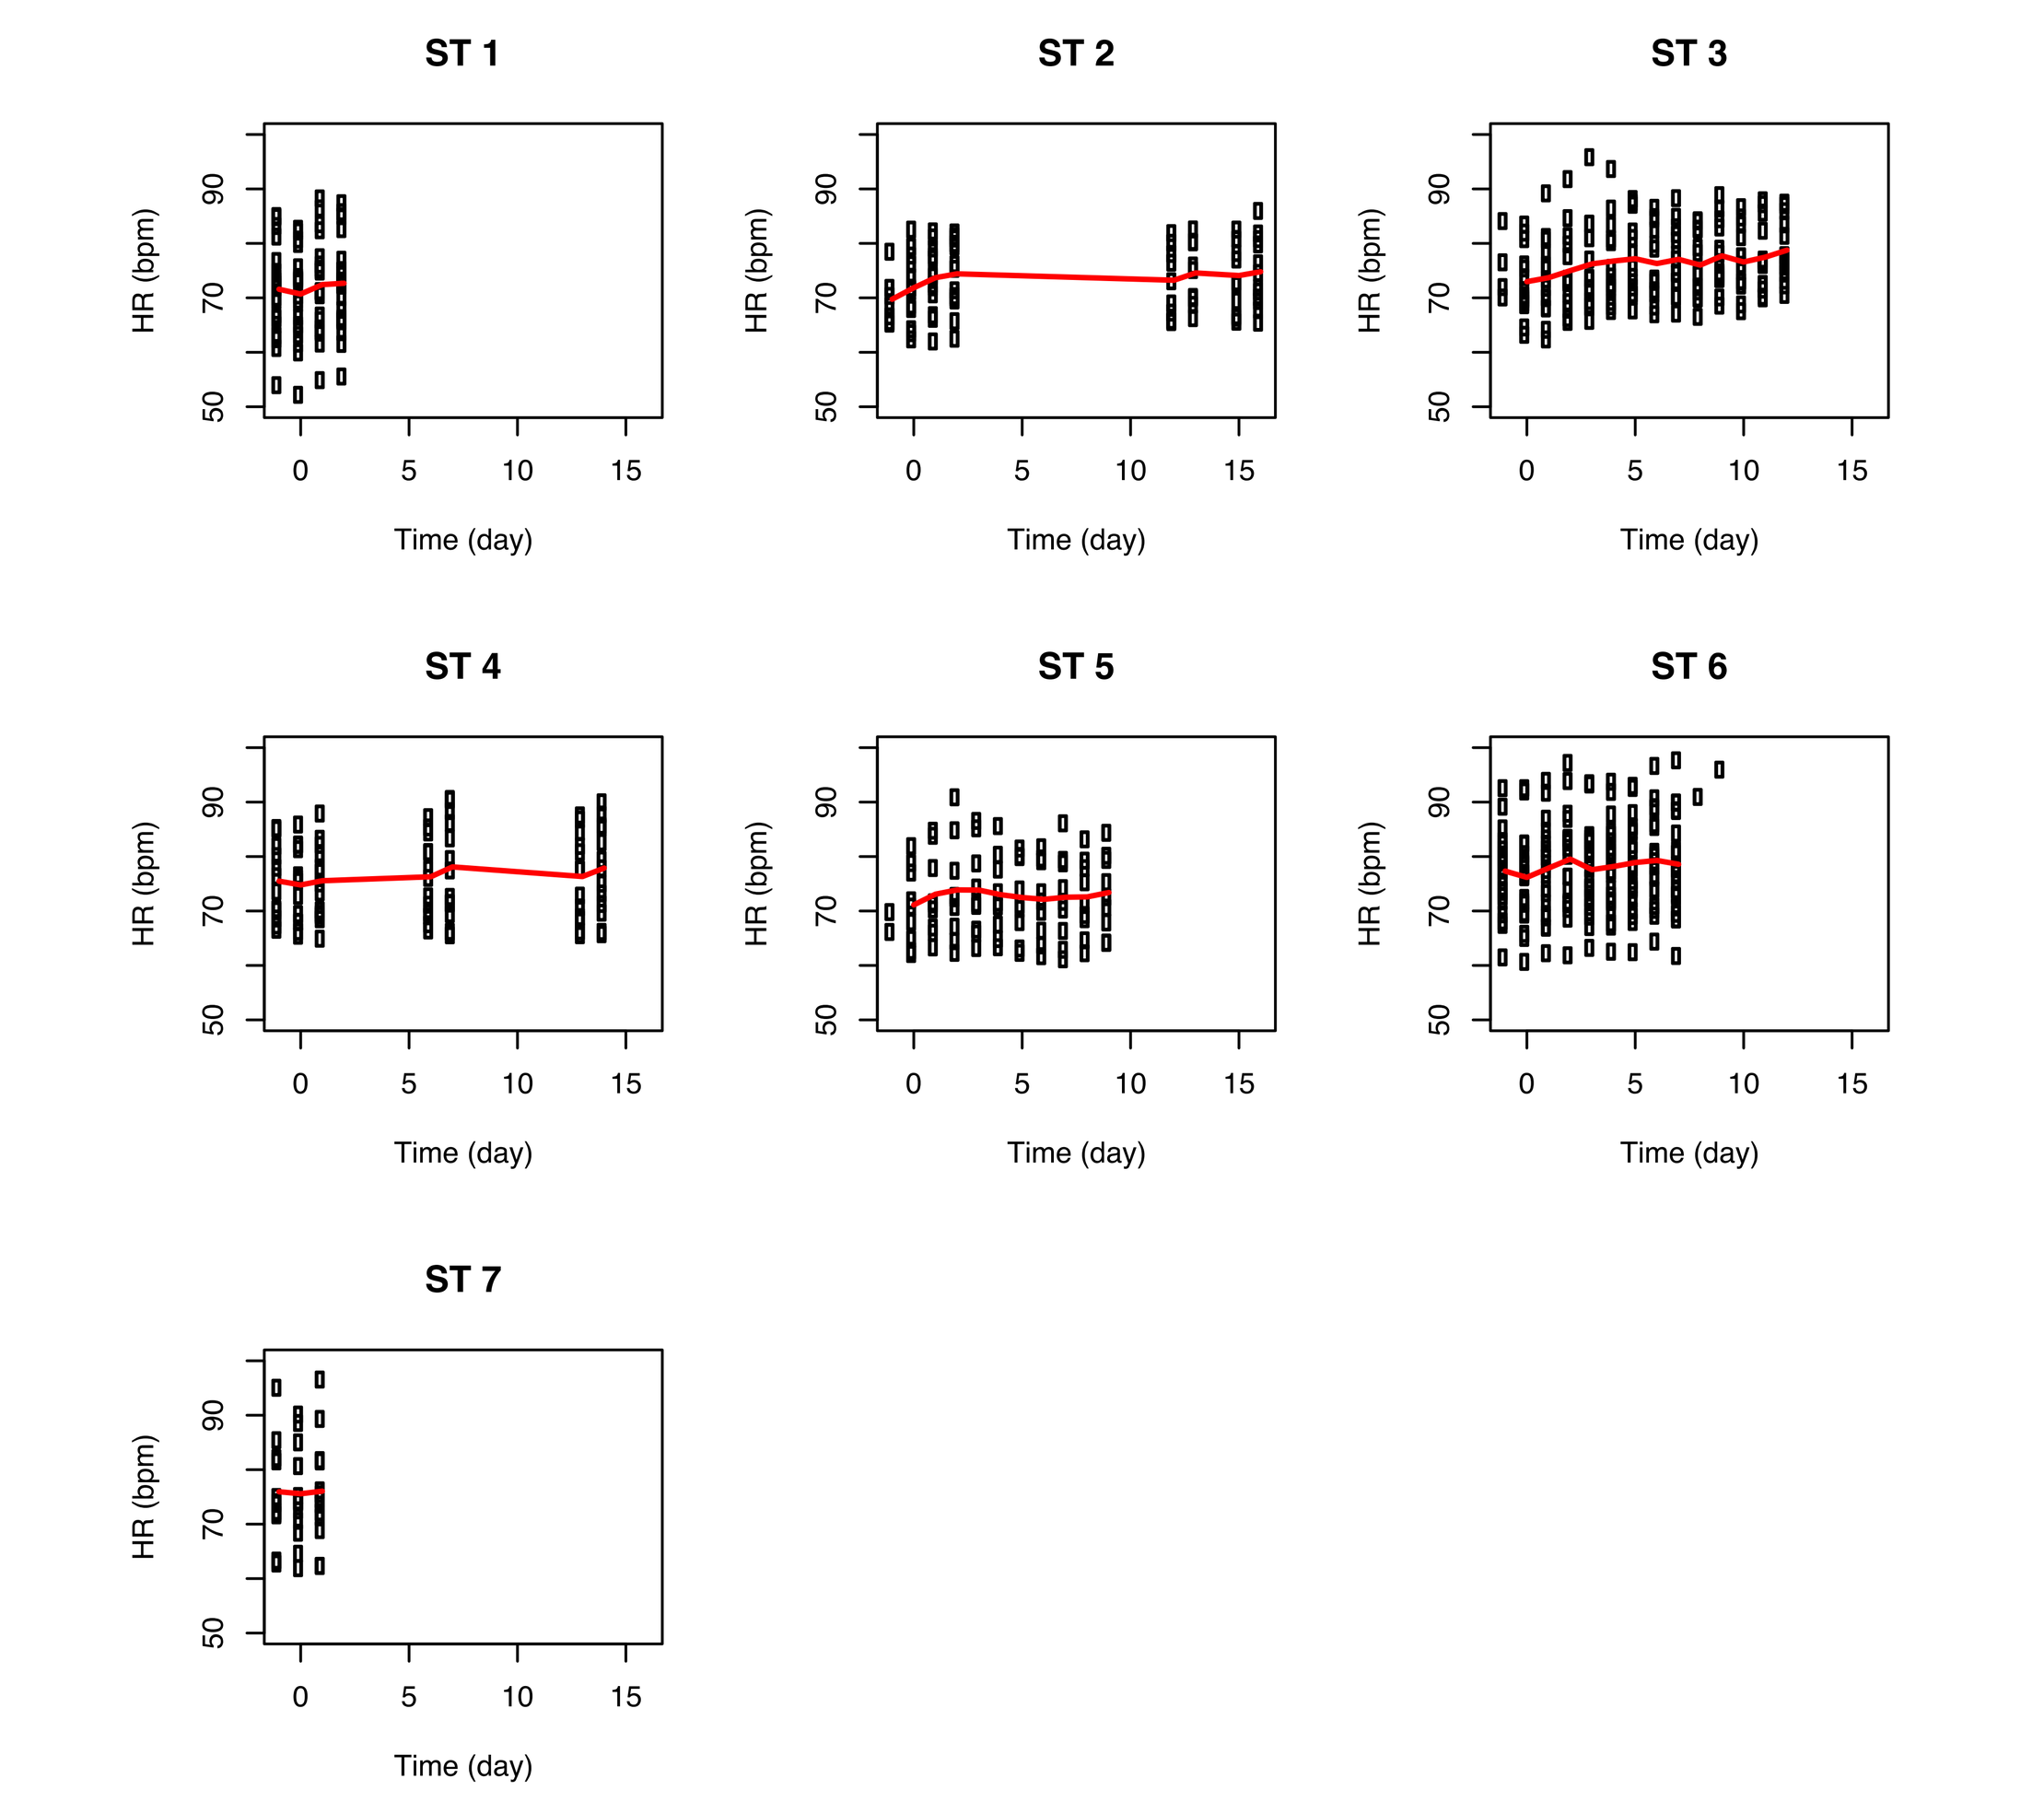
ST 1
ST 2
ST 3
ST 4
ST 5
ST 6
ST 7
HR (bpm)
Time (day)

IOV, Inter-occasion-variability

Conclusions

- Overall gender- and study-dependent effects were shown to influence the circadian changes of HR of the typical subject to a greater extent than what is considered to be a clinically relevant drug effect. However, not all the effects found to be statistically significantly different during model building could also be considered clinically relevant, especially a number of study-dependent effects on the amplitude parameters.
- Due to the high variability of HR over the day and the large study and gender dependencies, it is recommended to consider a model-based approach when estimating any potential drug effect compared to baseline and placebo during clinical trials.
- The hourly average used to collate the HR data provided a good estimate of the daily changes in HR, but is unable to describe short-term changes. Depending on the modeling objective, a finer level of granularity of the data (and thus a higher complexity of the model) might be more appropriate.

Acknowledgements

We want to thank Niclas Jonsson for his work on a similar project that inspired this work.

This study was supported by Novartis Pharma AG, Basel, Switzerland.
Copyright © 2010 Novartis Pharma AG, Basel, Switzerland.
All rights reserved.

References

1. Hempel G *et al.* Clin. *Pharm. Ther.* 1998; 64: 622–635.
2. Samson A *et al. Stat. Med.* 2007; 26: 4860–4875.
3. Jonsson EN & Karlsson MO. *Comput. Methods. Programs. Biomed.* 1999; **58**(1):51–64.
4. Beal S *et al* (eds.). *NONMEM User's Guides.* (1989-2006) Icon Development Solutions, Ellicott City, MD, USA, 2006.

Poster presented at the Population Approach Group Europe (PAGE), June 8–11, 2010, Berlin, Germany.